EXHANCE-12: A One Year Study of Safety and Efficacy of a Fluticasone Propionate Exhalation Delivery System (FLU-EDS), in Patients with Chronic Rhinosinusitis With and Without Polyps

James Palmer, MD[1]; Kraig Jacobson, MD[2]; John Messina, PharmD[3]; Colette Kosik-Gonzalez, BS, MA[3]; Per Djupesland, MD, PhD[3]; Ramy Mahmoud, MD, MPH[3]

1. Department of Otorhinolaryngology–Head and Neck Surgery, University of Pennsylvania, Perelman School of Medicine, Philadelphia, PA; 2. Oregon Allergy Associates, Eugene, OR; 3. OptiNose US Inc. Yardley, PA, USA and OptiNose AS, Oslo, Norway

BACKGROUND

- Chronic Rhinosinusitis with (CRSwNP) and without (CRSsNP) nasal polyps is common (11.5-27.9 million persons in the U.S.) and is associated with substantial disease burden. The overall annual economic burden of CRS in the U.S. was estimated at $22 billion (direct and indirect costs) in 2014.[1,2]

- Cardinal symptoms include nasal congestion/obstruction, rhinorrhea, facial pain/pressure, and reduction/loss of smell. Extrasinal manifestations are very common and include fatigue and bodily pain, sleep dysfunction, and depression. The overall effect is an impairment of quality of life (QoL) similar in degree to serious diseases such as CHF, COPD, and Parkinson's.[3,4]

- Inflammation is a hallmark of CRS and is generally present throughout the nasal/sinus cavities, inclusive of the ostiomeatal complex (OMC), where the sinus ostia drain and ventilate. Nasal polyps most commonly originate in this area.[5]

- Intranasal corticosteroid (INCS) sprays are part of standard of care in all treatment guidelines; however, conventional INCS are relatively inefficient and ineffective in delivery of topically-acting drug to key disease sites beyond the nasal valve area, including the OMC, potentially reducing treatment benefit, and are associated with low treatment satisfaction.[2]

- Exhalation Delivery Systems (EDS) **(Figure 1)** have been shown to deposit drug deeper and more broadly in the nasal cavity (particularly superiorly/posteriorly; e.g., OMC), with less drug loss to drip-out and swallowing, compared to conventional INCS. FLU-EDS (fluticasone propionate exhalation delivery system) uses a novel EDS to substantially modify delivery of locally-acting steroid (fluticasone), including to the OMC.[6,7]

- The objective of this study was to assess the long term safety and efficacy of FLU-EDS 372 µg twice daily (BID) in patients with moderate-severe CRS, with or without nasal polyps.

METHODS

- 52-week open-label, multicenter, repeated endoscopy study
 - Eligible patients were ≥18 years of age with CRSwNP/sNP, as determined by nasoendoscopy plus history of diagnostic symptoms for ≥12 weeks, and were currently experiencing ≥2 core symptoms of CRS, one of which had to be nasal congestion/obstruction or rhinorrhea.

Efficacy Assessments

- Lund-Mackay endoscopic assessment
- Nasal Polyp Grading Scale (CRSwNP patients)
- Sino-Nasal Outcome Test (SNOT-22)
- Surgical intervention assessment
- Medication evaluation questionnaire
- Patient Global Impression of Change (PGIC)

Safety Assessments

- Adverse Events (AEs)
- Nasal endoscopy
- Ocular examination by Ophthalmologist
- Clinical laboratory parameters
- Vital signs
- Physical examination

RESULTS

- 333 patients were screened, and 224 patients were enrolled at 21 centers in the U.S. 223 patients received study drug of which 144 (64.6%) completed the study.
- Baseline characteristics are shown in **Table 1**.

Table 1. Baseline Characteristics

Characteristic	CRSwNP (n=34)	CRSsNP (n=189)	Safety Set (n=223)
Age, mean (SD), y	46 (13.7)	45.3 (12.5)	45.4 (12.6)
Male sex, No. (%)	20 (58.8)	76 (40.2)	96 (43)
White race, No. (%)	30 (88.2)	145 (76.7)	175 (78.5)
Corticosteroids used for CRS in last 10 yrs, No. (%)	34 (100)	180 (95.2)	214 (96)
Bilateral endoscopic nasal polyp score, mean (SD)	2.8 (1.17)	-	-
Lund-Mackay total score, mean (SD)	1.7 (1.5)	1.4 (1.2)	1.4 (1.3)
SNOT-22 total score, mean (SD)	41.9 (23.3)	39.7 (21.7)	40 (21.9)
≥1 sinus surgery for polyp removal or sinus surgery, No. (%)	15 (44.1)	49 (25.9)	64 (28.7)

Fig 1. Exhalation Delivery System (EDS)[7]

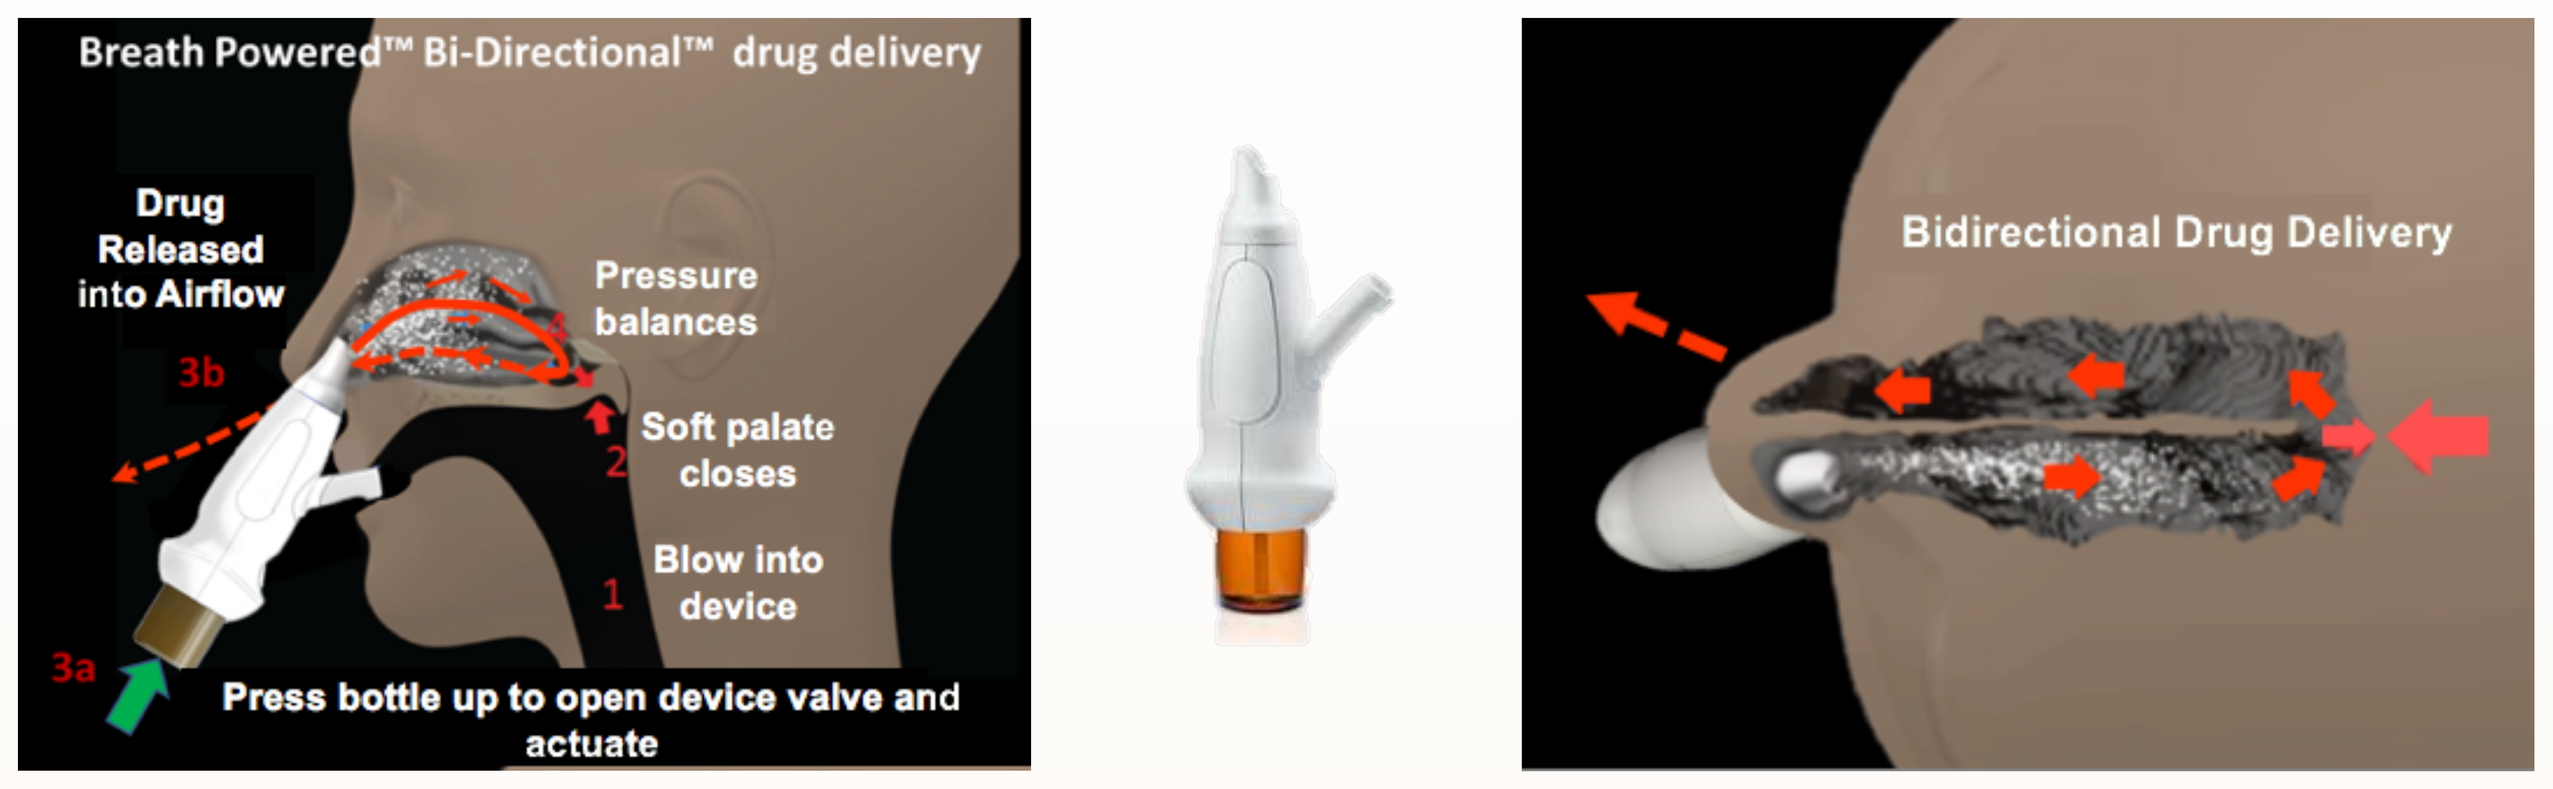
Breath Powered™ Bi-Directional™ drug delivery
Drug Released into Airflow
Pressure balances
Soft palate closes
Blow into device
Press bottle up to open device valve and actuate
1
2
3a
3b
Bidirectional Drug Delivery

- With FLU-EDS, mean SNOT-22 total scores decreased similarly in patients with and without nasal polyps (-19.4 and -18.6) and the improvement was maintained or gradually improving in both groups throughout the 12-month study. **(Figure 2)**

- In patients with and without nasal polyps, the median SNOT-22 at Month 12 was 8.5 and 9, respectively **(Figure 2)**. The average SNOT-22 scores among healthy volunteers has been reported to be 9.3.[8] Data from this study suggest that nearly half of patients treated with FLU-EDS for 12 months achieve SNOT-22 scores at or below those of healthy volunteers.

Fig 2. Mean and Median SNOT-22 Total Scores

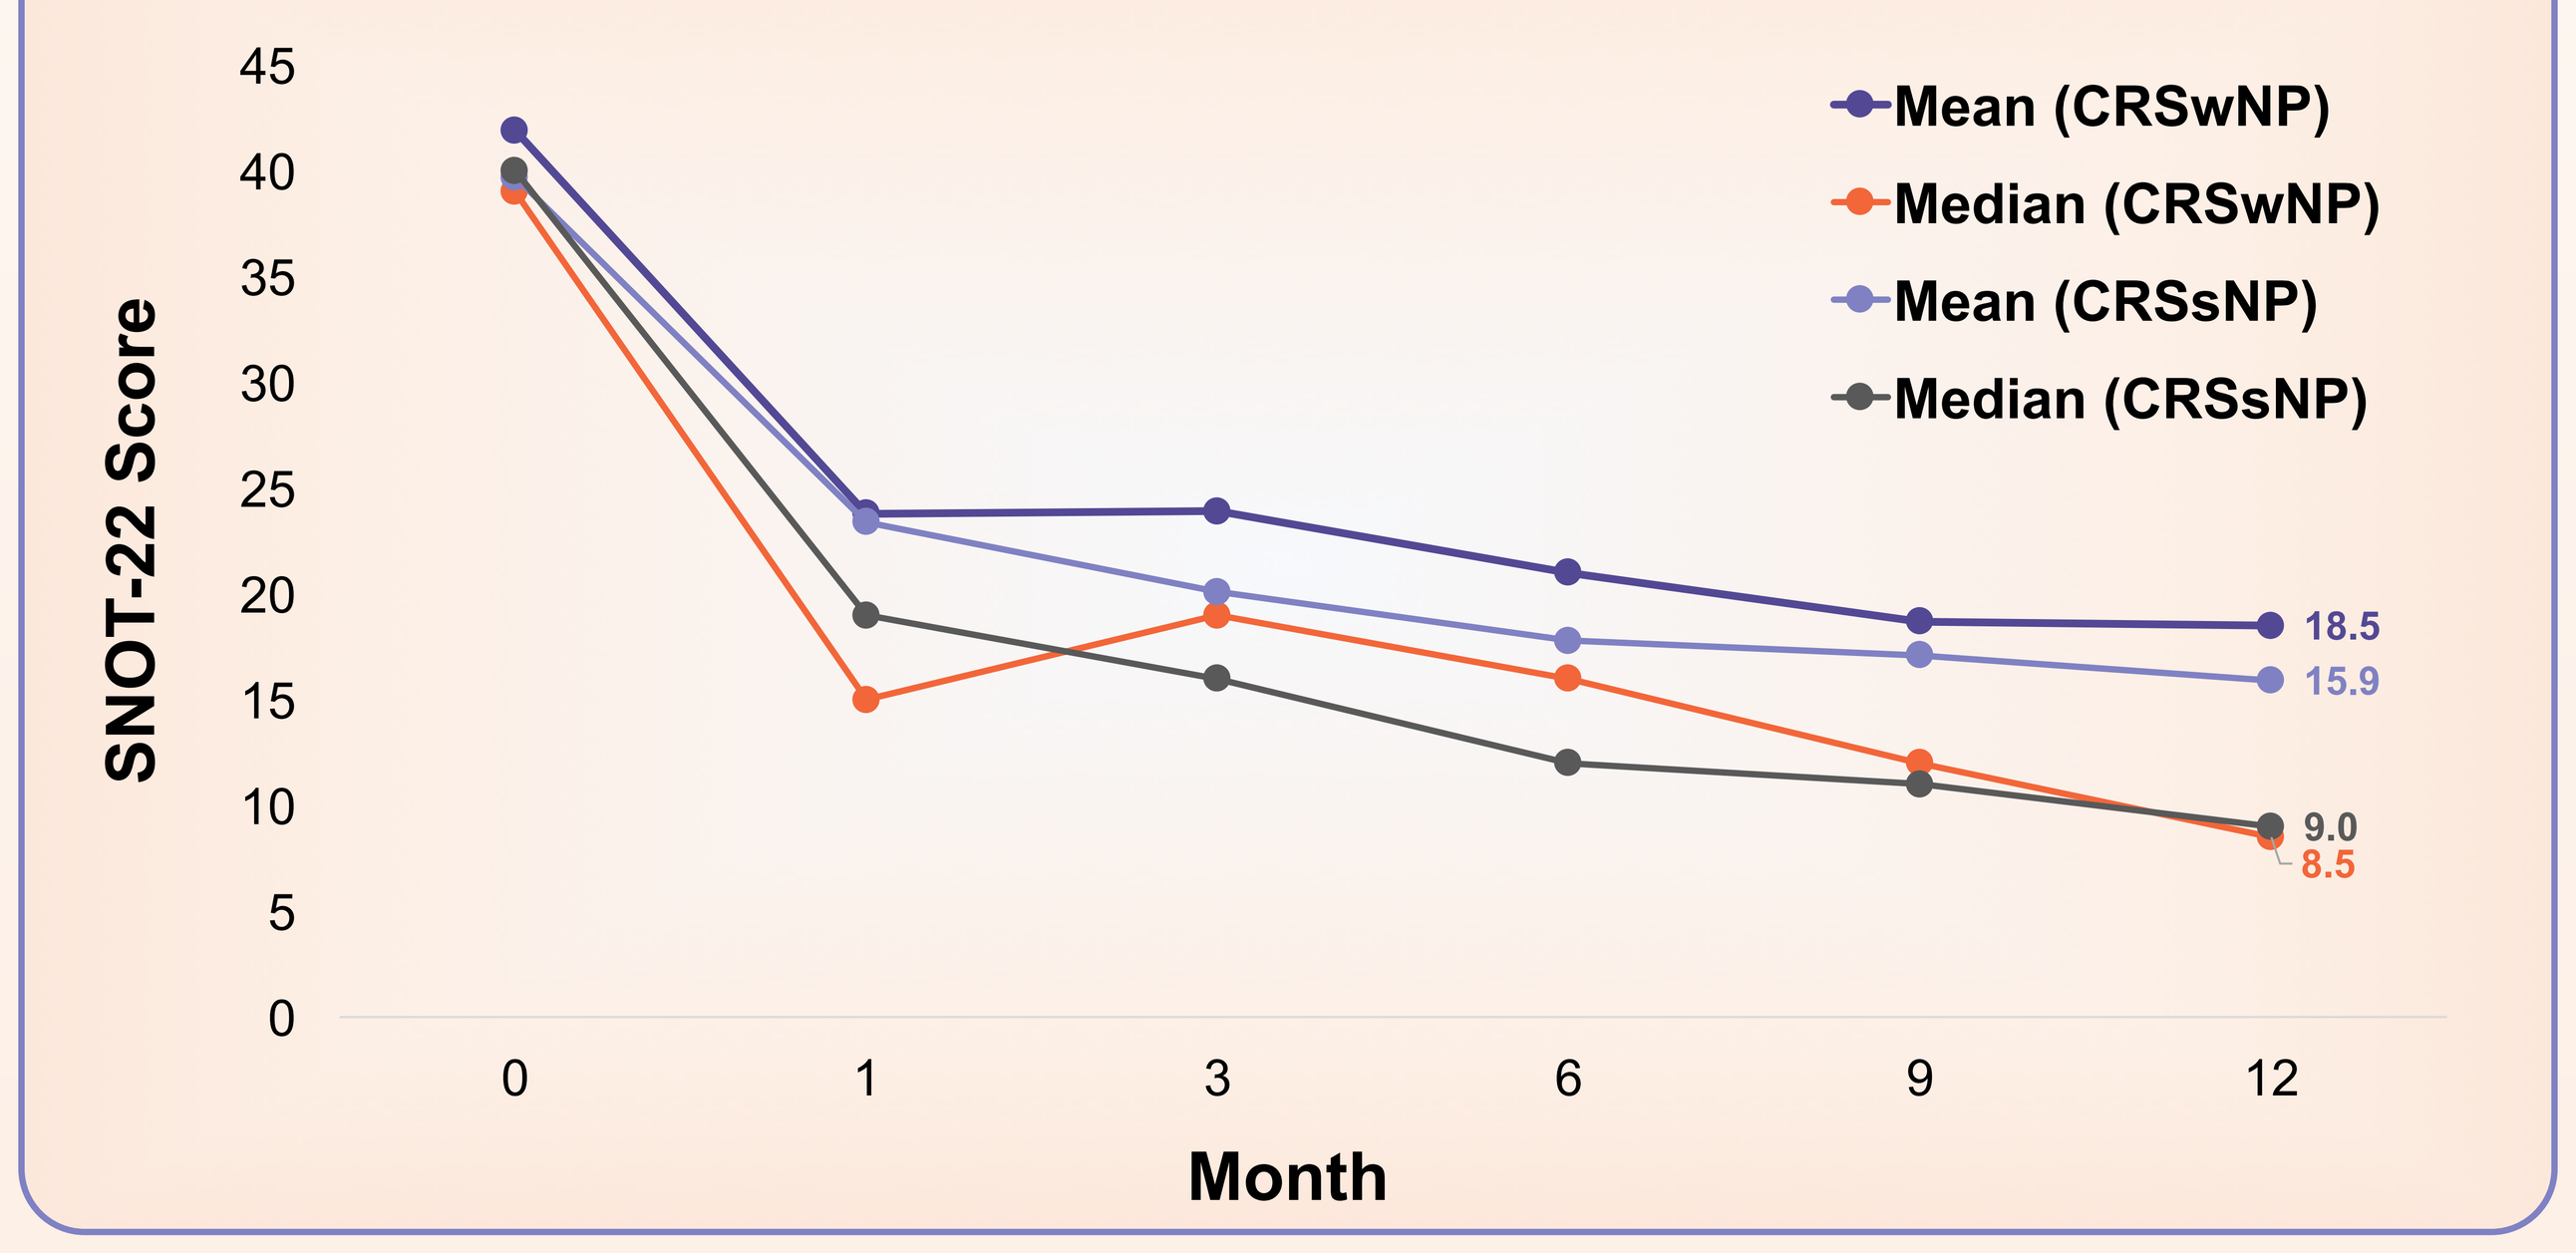
SNOT-22 Score
45
40
35
30
25
20
15
10
5
0
0
1
3
6
9
12
Month
Mean (CRSwNP)
Median (CRSwNP)
Mean (CRSsNP)
Median (CRSsNP)
18.5
15.9
9.0
8.5

- For patients with endoscopic Lund-Mackay edema scores >0 at baseline, FLU-EDS treatment was associated with complete resolution of edema in 50% of CRSwNP and 56% of CRSsNP patients at end-of-study. **(Figure 3)**

Fig 3. Patients with Complete Resolution of Edema

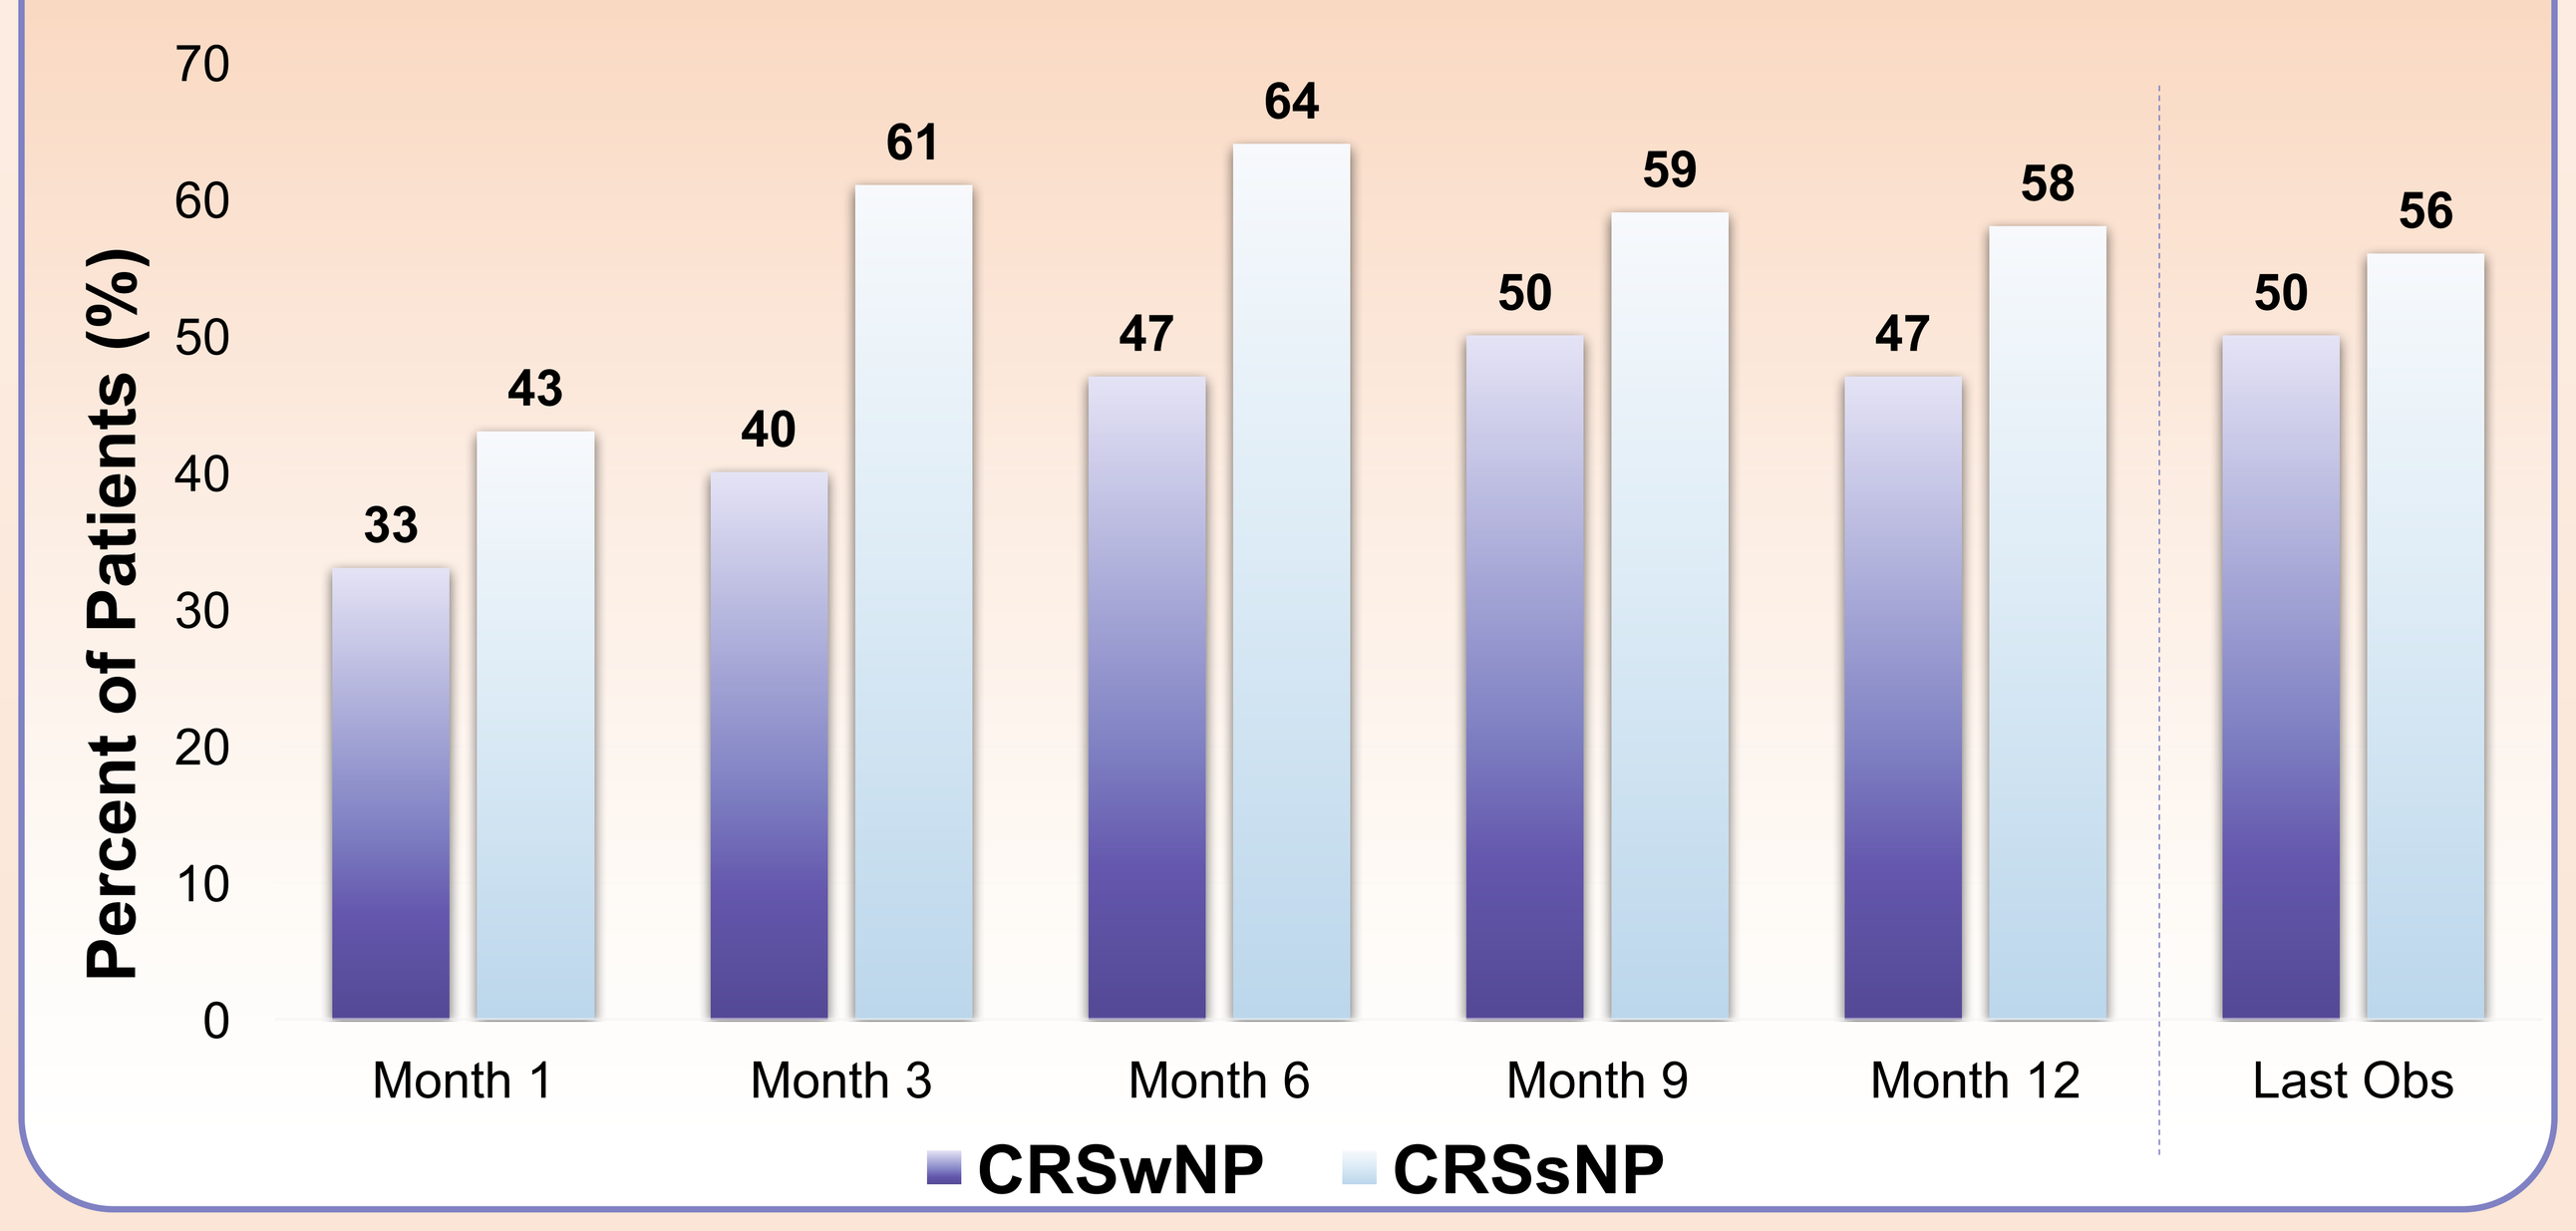
Percent of Patients (%)
70
60
50
40
30
20
10
0
33
43
40
61
47
64
50
59
47
58
50
56
Month 1
Month 3
Month 6
Month 9
Month 12
Last Obs
CRSwNP
CRSsNP

- For CRSwNP, the percentage observed endoscopically to have polyp elimination in at least one nostril increased steadily through 12 months. Among those completing 12 months of treatment, 54.2% had polyp elimination in at least one nostril. **(Figure 4)**

Fig 4. CRSwNP Patients with Polyps Eliminated in ≥ 1 Nostril

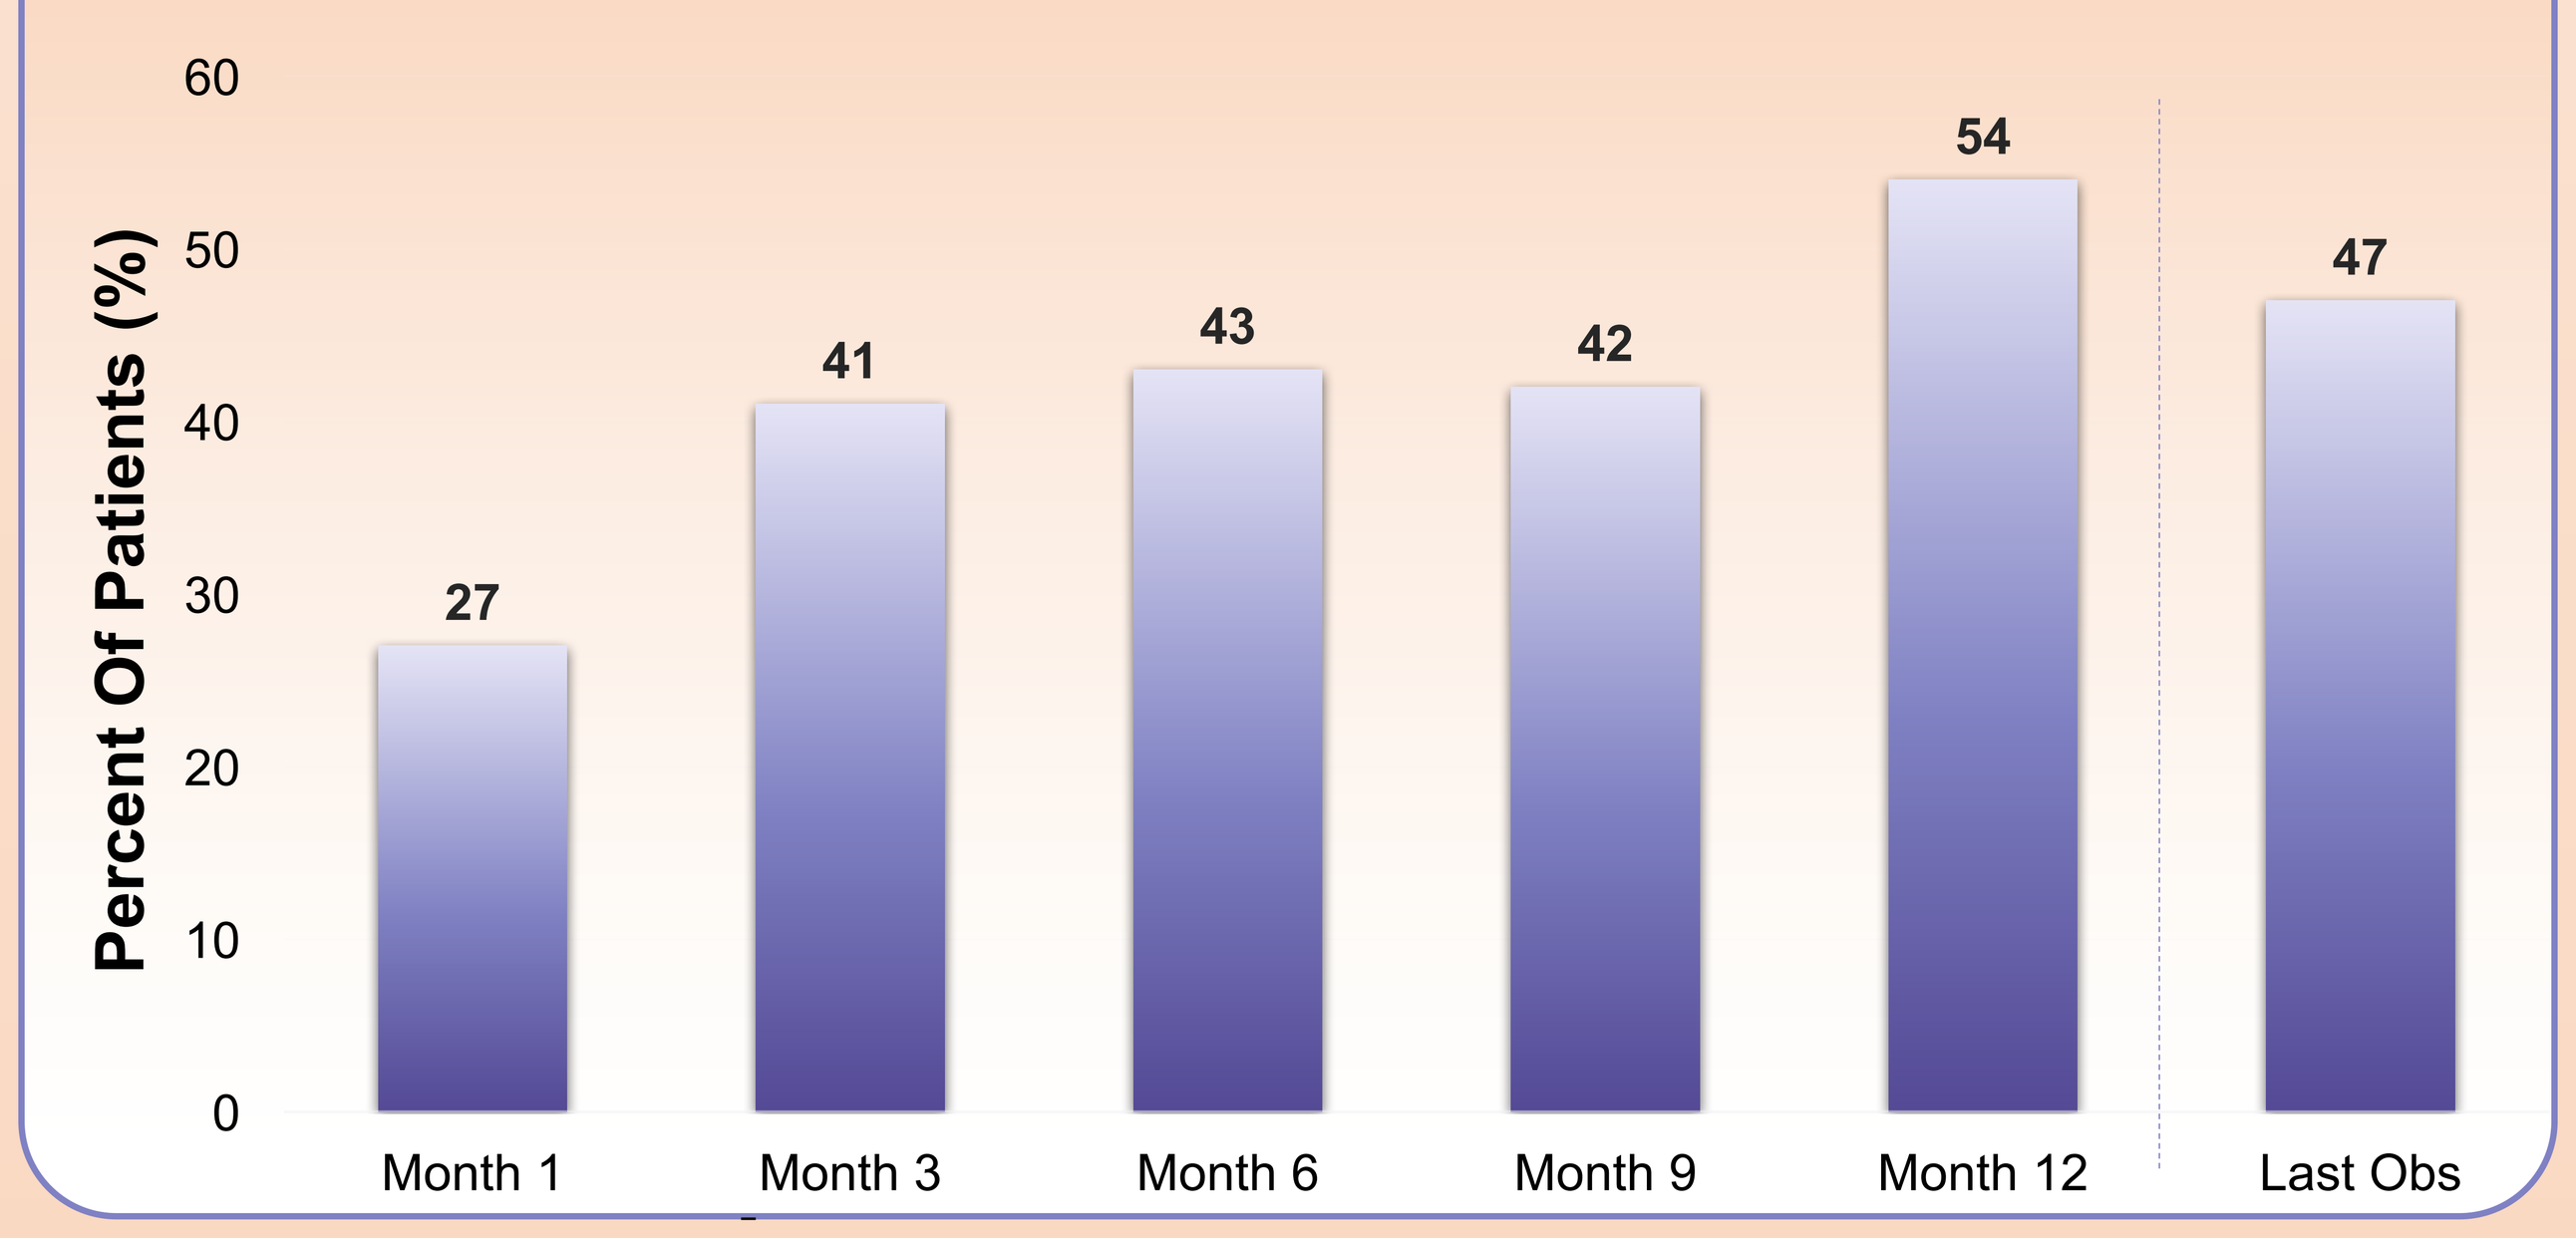
Percent Of Patients (%)
60
50
40
30
20
10
0
27
41
43
42
54
47
Month 1
Month 3
Month 6
Month 9
Month 12
Last Obs

- Following 1 month of FLU-EDS treatment, 54.5% of patients with nasal polyps experienced at least one point improvement in polyp grade. This increased to 83.3%, after 12 months of treatment. **(Figure 5)**

Fig 5. CRSwNP Patients with ≥ 1 Point Change in Polyp Grade

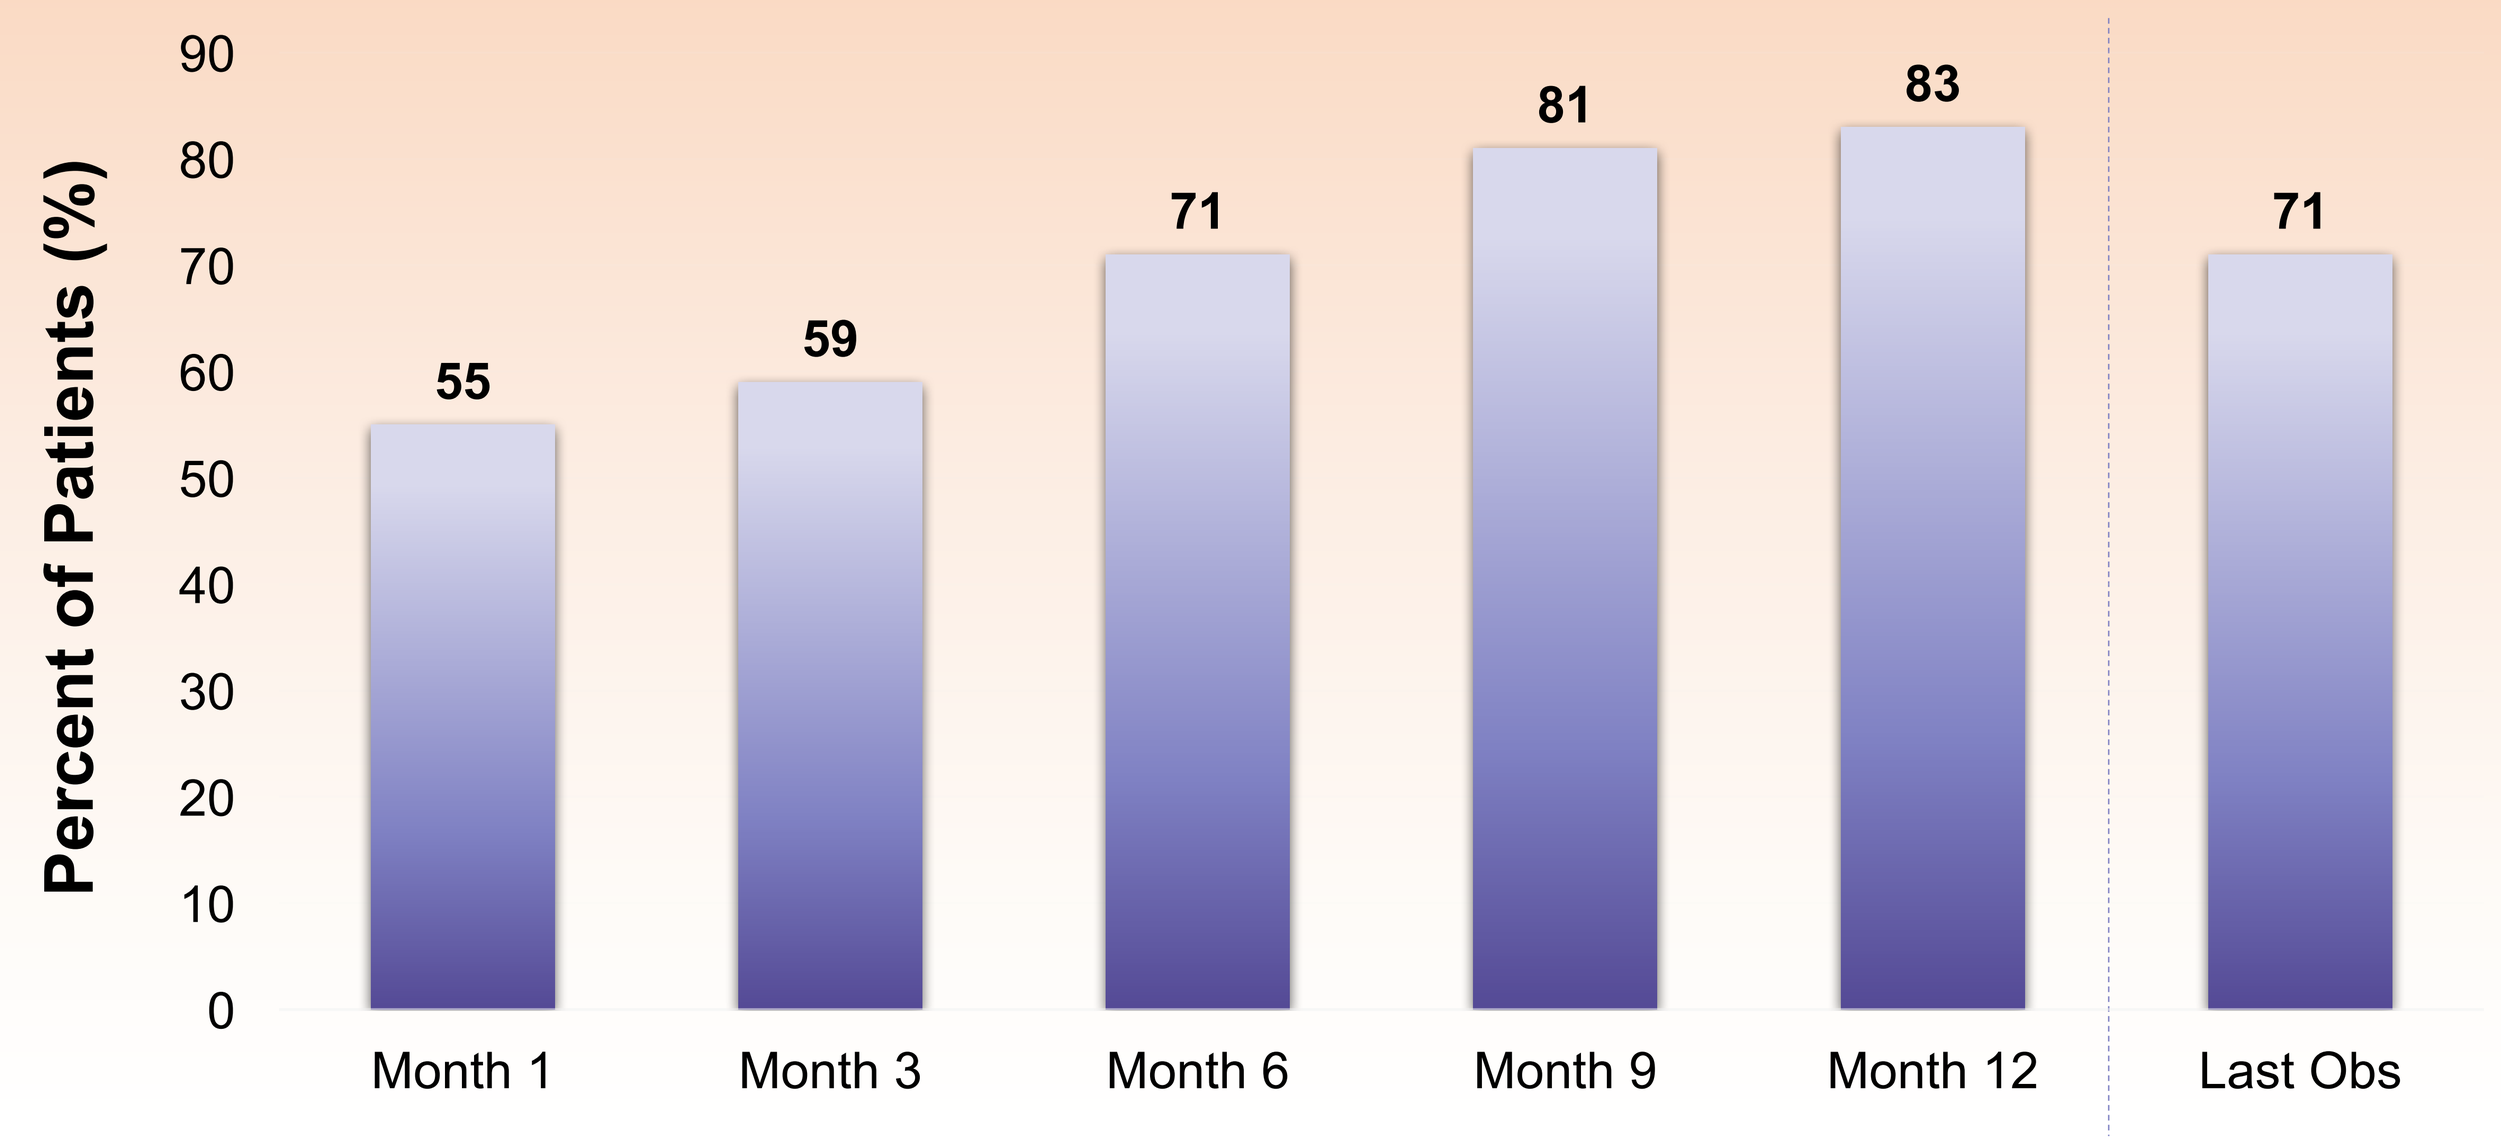
Percent of Patients (%)
90
80
70
60
50
40
30
20
10
0
55
59
71
81
83
71
Month 1
Month 3
Month 6
Month 9
Month 12
Last Obs

- Global Improvement: Increasingly through 12 months, patients with and without nasal polyps using FLU-EDS reported global improvement in symptoms. At end-of-study, 87% of patients reported improvement, with 72.1% reporting 'much' or 'very much' improvement.

- Ease of Use: 187 of 218 patients (85.8%) reported FLU-EDS was 'easy' or 'somewhat easy' to use. 161 patients (73.9%) reported that it was 'somewhat' or 'very' comfortable.

- Undesired Loss of Drug (compared to conventional INCS): A total of 125 patients (57.3%) reported 'less' or 'much less' drip out of their nose; 155 patients (71.1%) reported 'less' or 'much less' drip down the back of the throat with FLU-EDS compared to their pre-study conventional INCS spray.

- By end-of-study, the number of patients with nasal polyps who qualified for surgical intervention decreased from 47.1% to 14.7% with continued use of FLU-EDS.

- FLU-EDS was well tolerated, with an adverse event (AE) profile similar to that reported with conventional INCS sprays in studies of comparable duration.

- The majority of AEs were mild in severity and diminished with continued use of study drug. The most common AEs (>5%) included epistaxis (27.8%, all types, spontaneously reported and identified on nasal exam), nasal mucosal disorder (17.5%), nasal septum disorder (14.3%), acute sinusitis (13.9%), nasal septal ulceration (11.2%), headache (9.4%), and upper respiratory infection (7.6%).

- There was no evidence of increasing intraocular pressure (IOP) or subcapsular cataracts.

CONCLUSIONS

- FLU-EDS 372 µg BID substantially improved subjective symptoms and objective local signs of disease with a magnitude of improvement similar in CRS patients with and without nasal polyps.

- Patients with nasal polyps at baseline experienced extensive reduction in polyp grade that continued to improve throughout the follow-up period.

- The AEs reported were generally local in nature, tended to be mild in severity, and were largely transient. AEs did not increase in frequency or severity with increasing duration of exposure to study drug.

- In patients with moderate to severe CRS (with and without polyps) and frequent past use of conventional INCS sprays, FLU-EDS was well tolerated and significantly improved symptoms and objective measures of disease, indicating the important role of superior/posterior drug delivery via EDS.

References:
1. Smith KA, Orlandi RR and Rudmik L. Cost of adult chronic rhinosinusitis: A systematic review. *Laryngoscope* 2015;125:1547-1556
2. Palmer J, Messina J, Biltech R, Grosel K, Mahmoud R. A Cross-Sectional Population-Based Survey of the Prevalence, Disease Burden, and Characteristics of the US Adult Population with Symptoms of Chronic Rhinosinusitis (CRS). Poster session presented at: the 62nd Annual Meeting of the American Rhinologic Society; 2016 Sept 16-17; San Diego, CA.
3. Orlandi RR, Kingdom TT, Hwang PH, et al. International Consensus Statement on Allergy and Rhinology: Rhinosinusitis. Int Forum. *Allergy Rhinol* 2016;6:S22-S209.
4. Soler ZM, Wittenberg E, Schlosser RJ, et al. Health state utility values in patients under- going endoscopic sinus surgery. *Laryngoscope* 2011;121:2672–2678.
5. Larsen PL, Tos M. Origin of nasal polyps: an endoscopic autopsy study. *Laryngoscope* 2004;114:710-719.
6. Djupesland PG, Messina J, Mahmoud R. Novel Nasal Exhalation Delivery Systems (EDS) May Produce Beneficial Activity Independent of Delivered Drug in Inflammatory Nasal Diseases and Migraine via Exhaled CO2 and Mucosal pH Changes. Poster session presented at: the 62nd Annual Meeting of the American Rhinologic Society; 2016 Sept 16-17; San Diego, CA
7. Djupesland PG. Nasal drug delivery devices: characteristics and performance in a clinical perspective – a review. *Drug Deliv and Transl Res. 2013;3:42-62*
8. Gillett S, Hopkins C, Slack R, et al. A pilot study of the SNOT 22 score in adults with no sinonasal disease. *Clin Otolaryngol.* 2009;34:467-469

Contact: John Messina, PharmD, OptiNose US Inc. Yardley, PA, USA.
Address: 1010 Stony Hill Rd., Yardley, PA, 19067
Email: john.messina@optinose.com, Phone: (267) 364-3500